Novel snake venom-based hemocoagulase bypasses coagulation to enhance hemostasis and limit bleeding in murine model of hemophilia A

Nicole Rhoads[1], Livia Stanger[2], Michael Wong[3], Benjamin Xiaoyi Li[3], Reheman Adili[1]

[1] Bloodworks Research Institute, Seattle, WA; [2]Department of Pharmacology, University of Michigan, Ann Arbor, MI; [3]Lee's Pharmaceutical, Shatin, Hong Kong

INTRODUCTION

Hemophilia A is a hereditary bleeding disorder and FVIII replacement has been the mainstay of treatment. However, there is an unmet need for novel therapeutics to prevent and treat bleeding. We recently reported that slounase, a thrombin-like enzyme batroxobin from a snake venom containing Factor X activator, enhances platelet-fibrin clot formation in heparin-anticoagulated mice, suggesting slounase may bypass coagulation to restore hemostasis. The effect of slounase on hemostasis and bleeding was determined in FVIII$^{-/-}$ mice, with and without inhibitors, using intravital microscopy hemostatic models, thromboelastography and bleeding assays.

METHODS

- Laser-induced cremaster arteriole thrombosis model, with and without inhibitors
- Saphenous vein hemostasis model
- $FeCl_3$-induced carotid artery thrombosis model
- Tail bleeding and hepatic bleeding models
- Thromboelastography using blood from FVIII$^{-/-}$ mice

RESULTS

FVIII$^{-/-}$ mice are unable to form hemostatic clots due to a severe defect in platelet accumulation and absence of fibrin formation at the site of vascular injury, as confirmed in hemostatic models under real-time intravital microscopy. Prophylactic intravenous treatment of 1U/kg slounase in FVIII$^{-/-}$ mice significantly enhanced platelet activation, accumulation, and fibrin formation in response to vascular injury resulting in stable hemostatic clot formation in the cremaster artery and saphenous vein laser ablation hemostasis models. Platelet-fibrin hemostatic clots also formed following ferric chloride injury to the carotid artery in FVIII$^{-/-}$ mice pretreated with slounase without reaching vessel occlusion. Importantly, in vivo hemostatic enhancement persisted in FVIII$^{-/-}$ mice intravenously treated with anti-FVIII antibodies and slounase. Furthermore, the hemostatic effect of slounase was confirmed by in vitro thromboelastography and in vivo bleeding assays.

ACKNOWLEDGEMENTS

This work was funded by a grant from Lee's Pharmaceutical Holdings Limited to RA.

SLOUNASE TREATMENT ENHANCES HEMOSTATIC CLOT FORMATION IN FVIII$^{-/-}$ MICE *IN VIVO*

Laser-induced cremaster arteriole thrombosis model

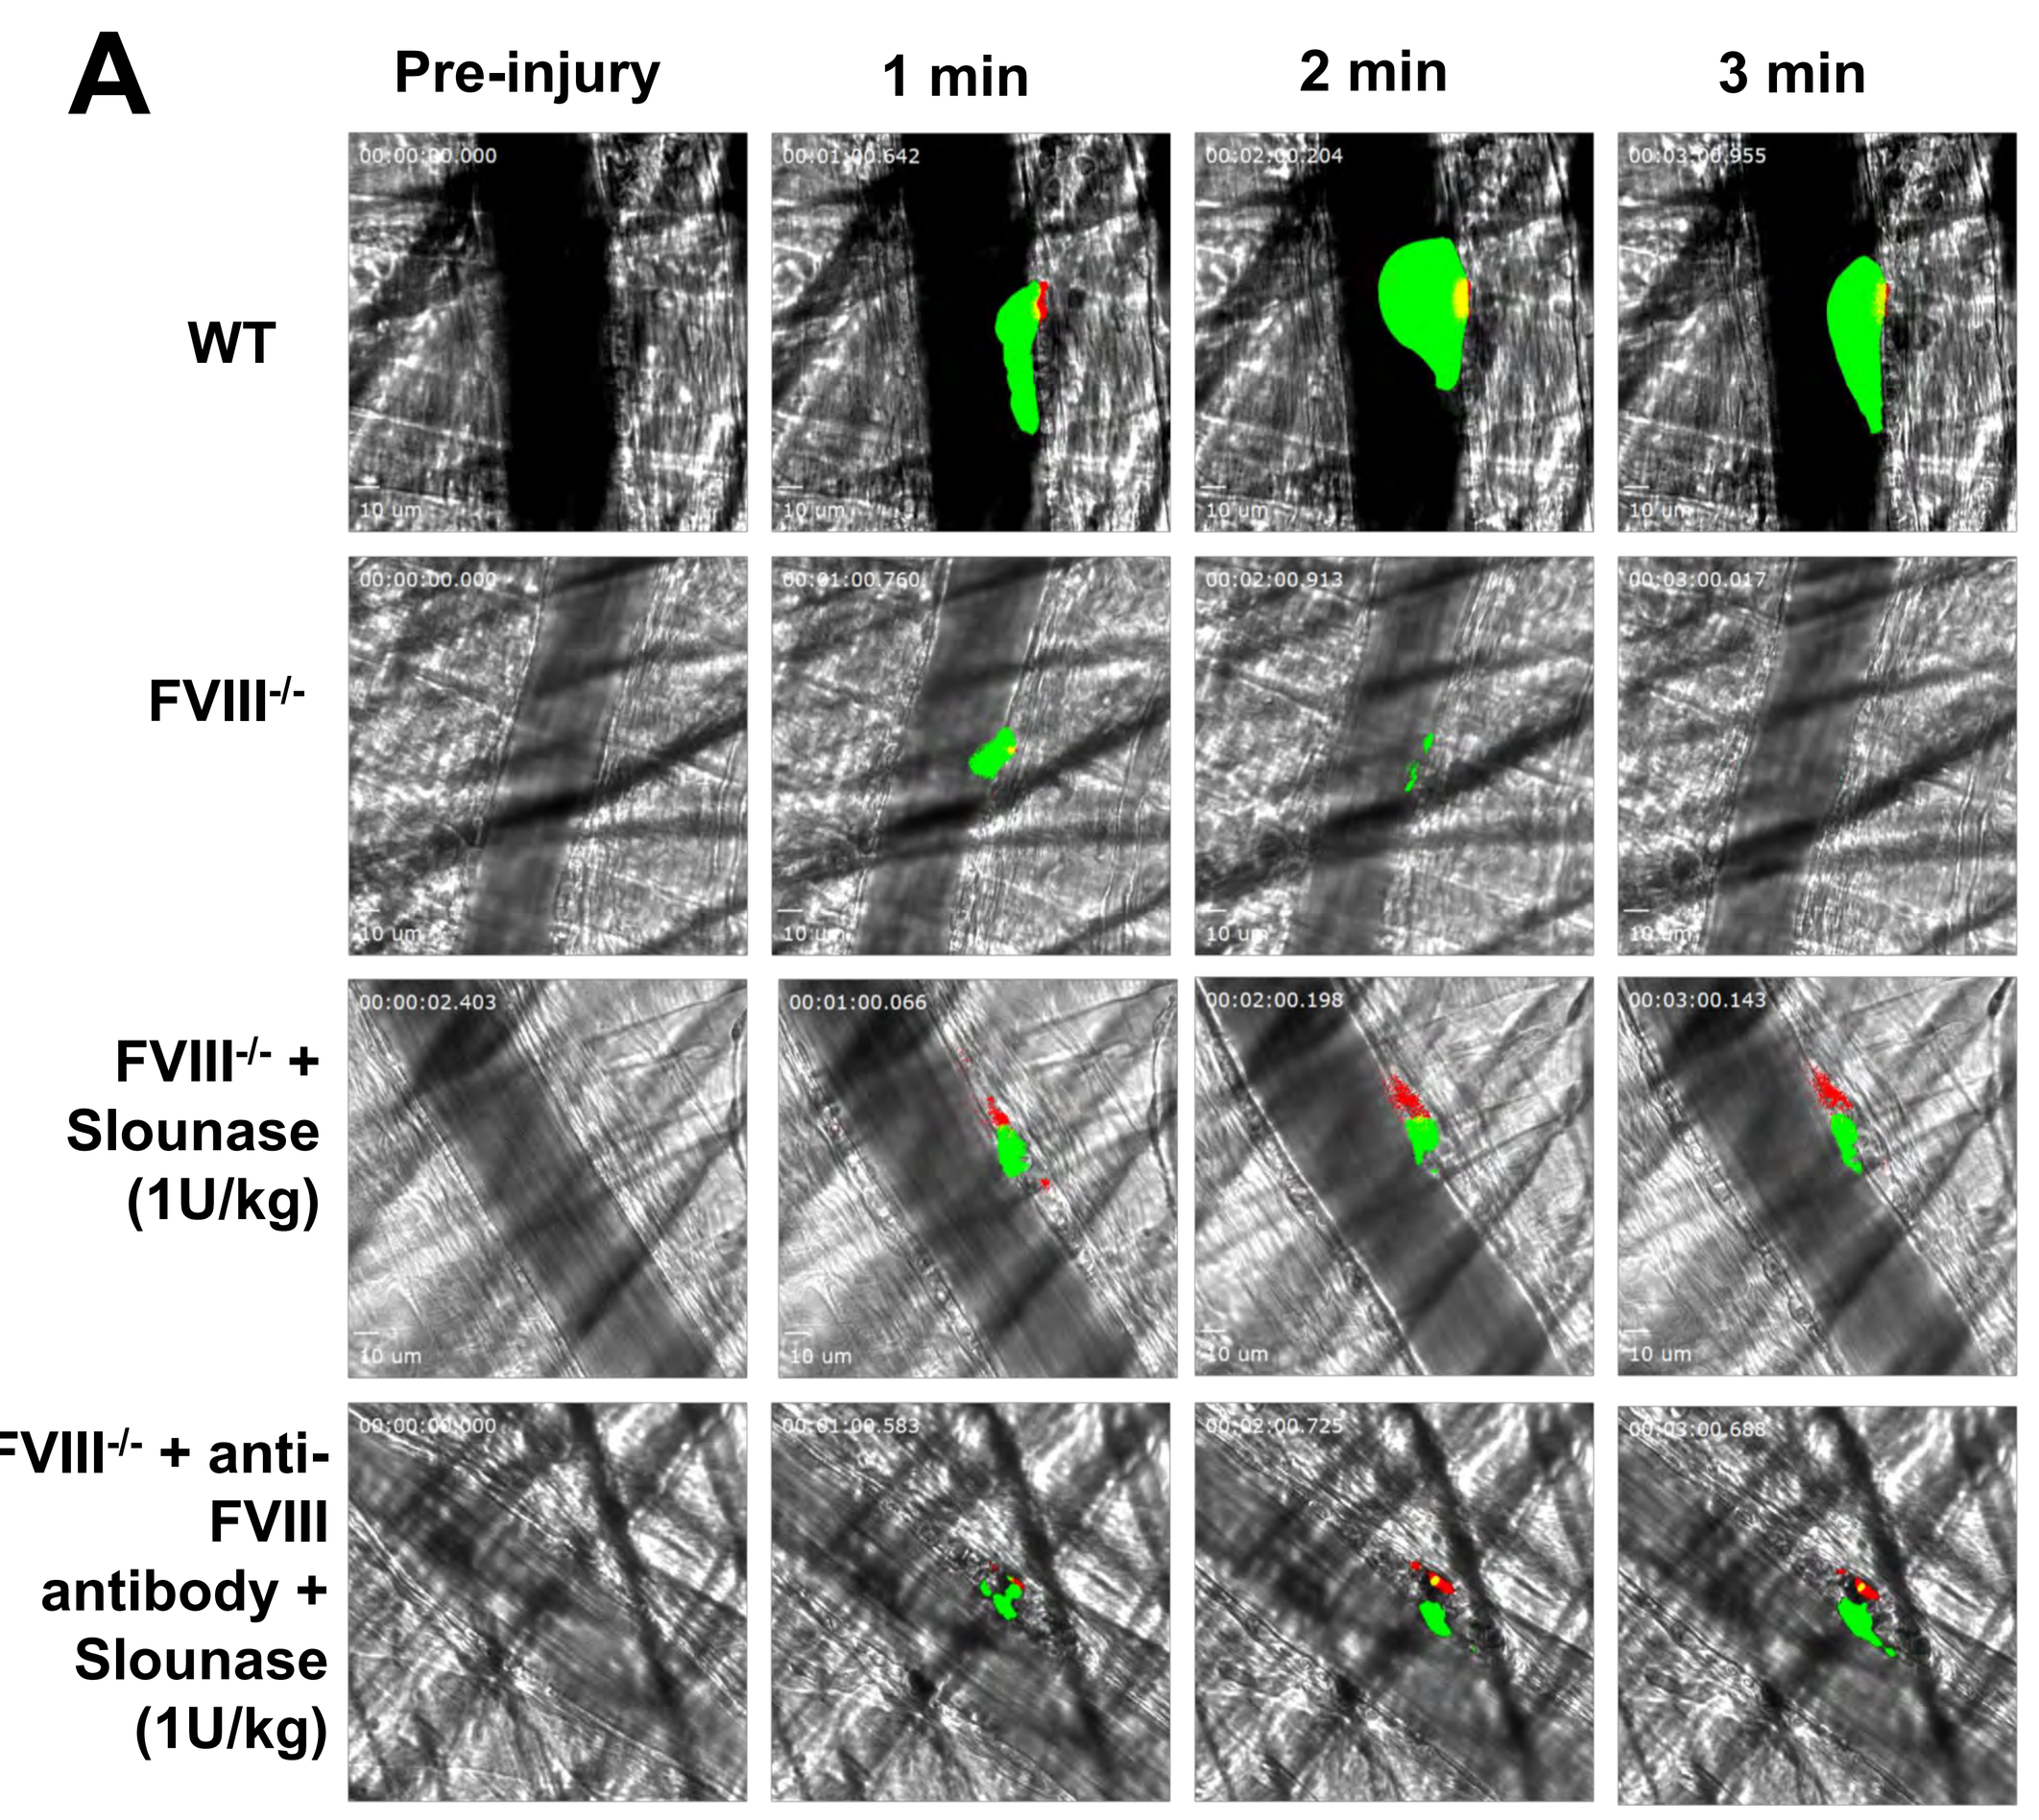
A
Pre-injury
1 min
2 min
3 min
WT
FVIII-/-
FVIII-/- + Slounase (1U/kg)
FVIII-/- + anti-FVIII antibody + Slounase (1U/kg)

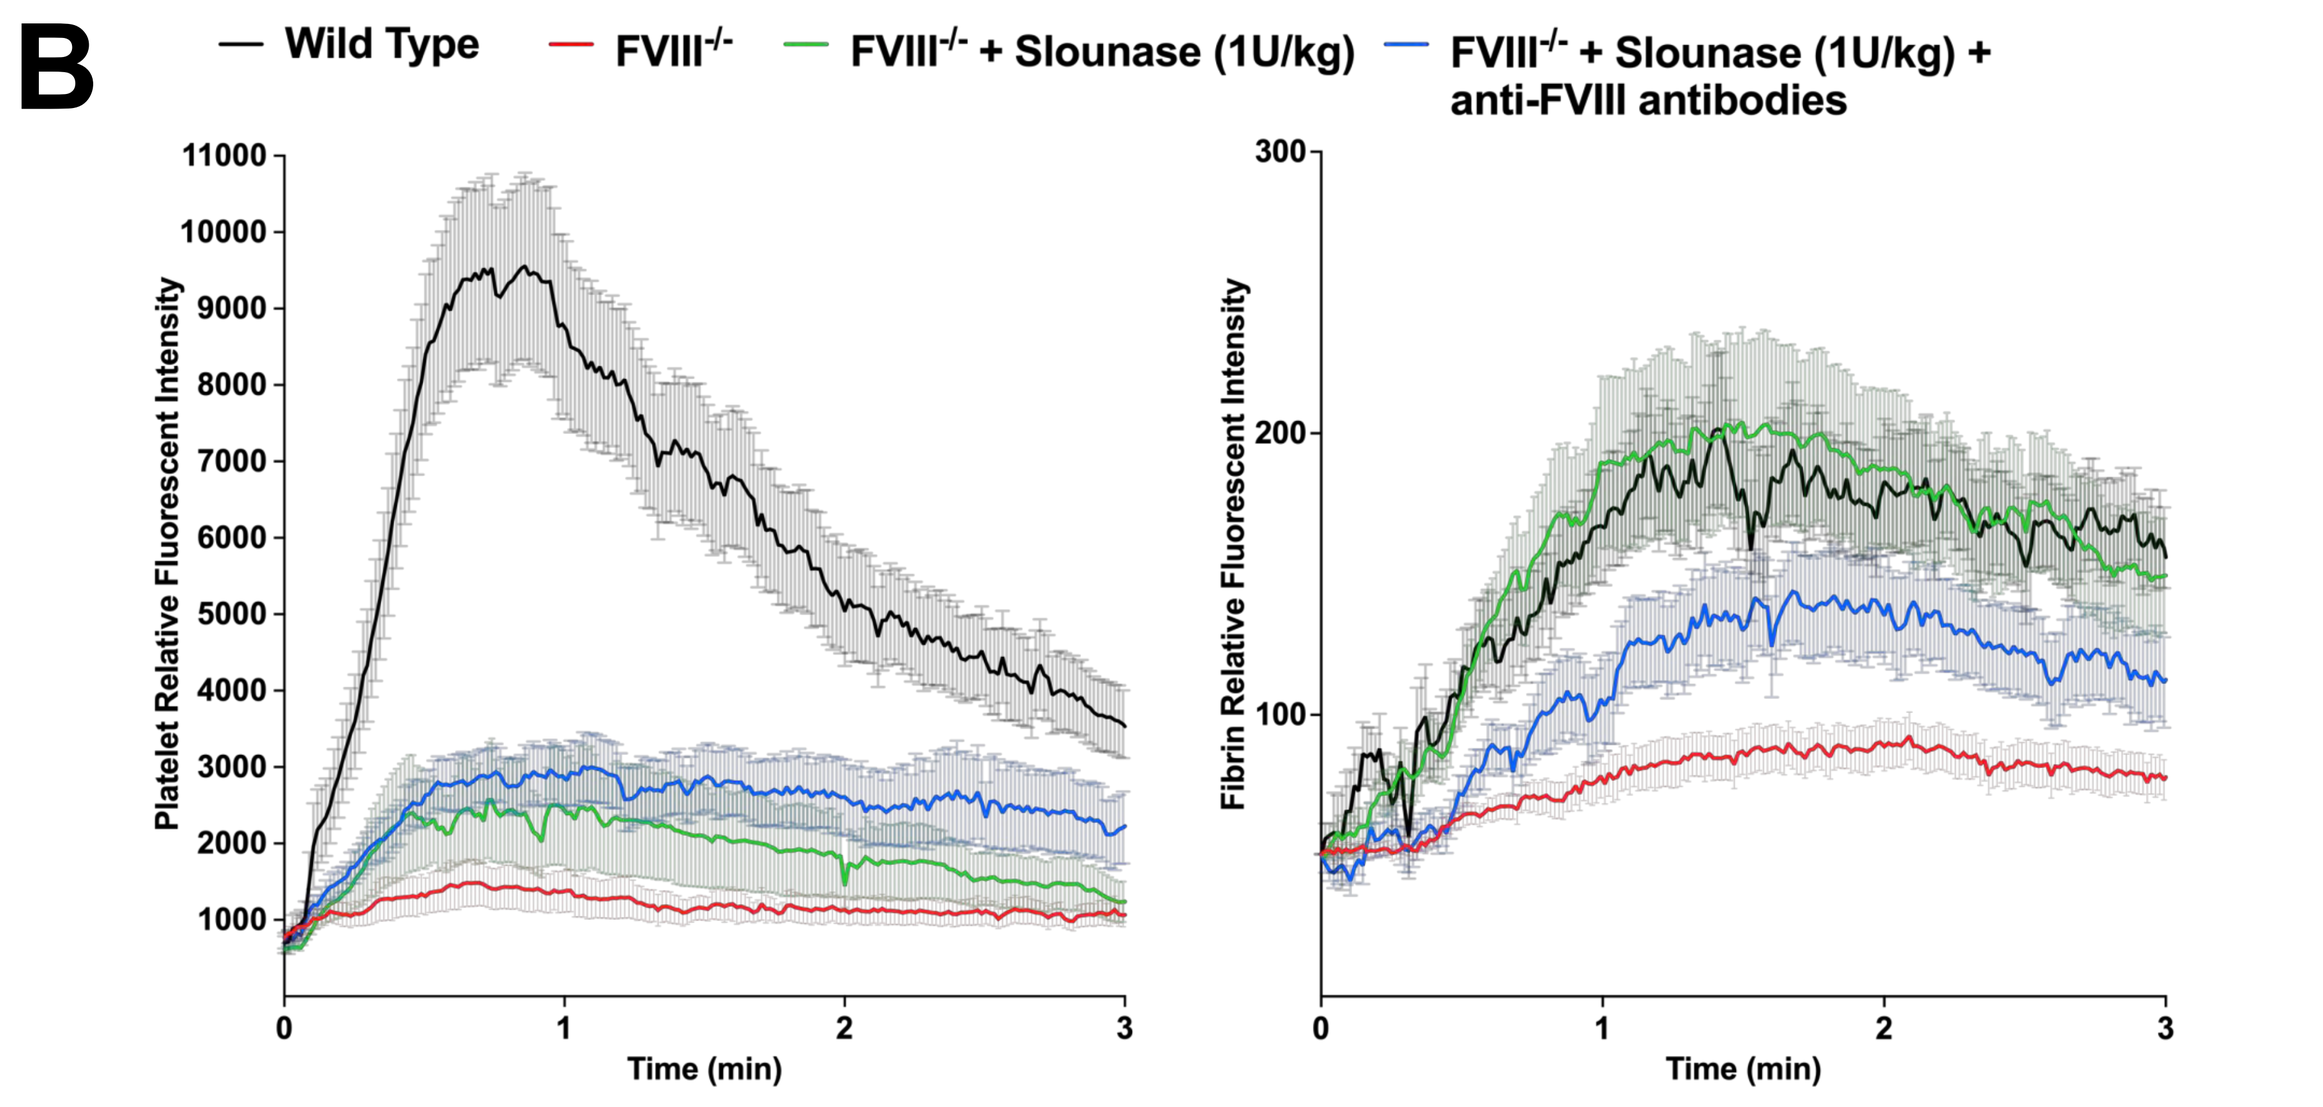
B
Wild Type
FVIII-/-
FVIII-/- + Slounase (1U/kg)
FVIII-/- + Slounase (1U/kg) + anti-FVIII antibodies
Platelet Relative Fluorescent Intensity
Fibrin Relative Fluorescent Intensity
Time (min)

Figure 1: Thrombosis in microcirculation of FVIII$^{-/-}$ mice is severely defective compared to WT controls and prophylactic slounase treatment enhanced thrombus formation in FVIII$^{-/-}$ mice with or without the presence of anti-FVIII antibodies. (A) Representative pictures of platelet accumulation (green) and fibrin formation (red) within the growing thrombus following laser injury in cremaster arteriole recorded in real time under an intravital microscope. Thrombotic response to vascular injury is severely impaired in FVIII$^{-/-}$ mice compared to WT controls as assessed by platelet accumulation and fibrin formation at the site of vascular injury. Intravenous treatment of slounase (1U/kg) in FVIII$^{-/-}$ mice notably enhanced both platelet accumulation and fibrin formation and resulted in a stable hemostatic clot formed at the site of vascular injury. The ability of slounase to restore *in vivo* hemostasis in FVIII$^{-/-}$ mice was not affected by the presence of circulating anti-FVIII antibodies. **(B)** Quantitative analysis of dynamics of platelet recruitment (left) and formation of fibrin (right) in the thrombus over time.

$FeCl_3$-induced carotid artery thrombosis model

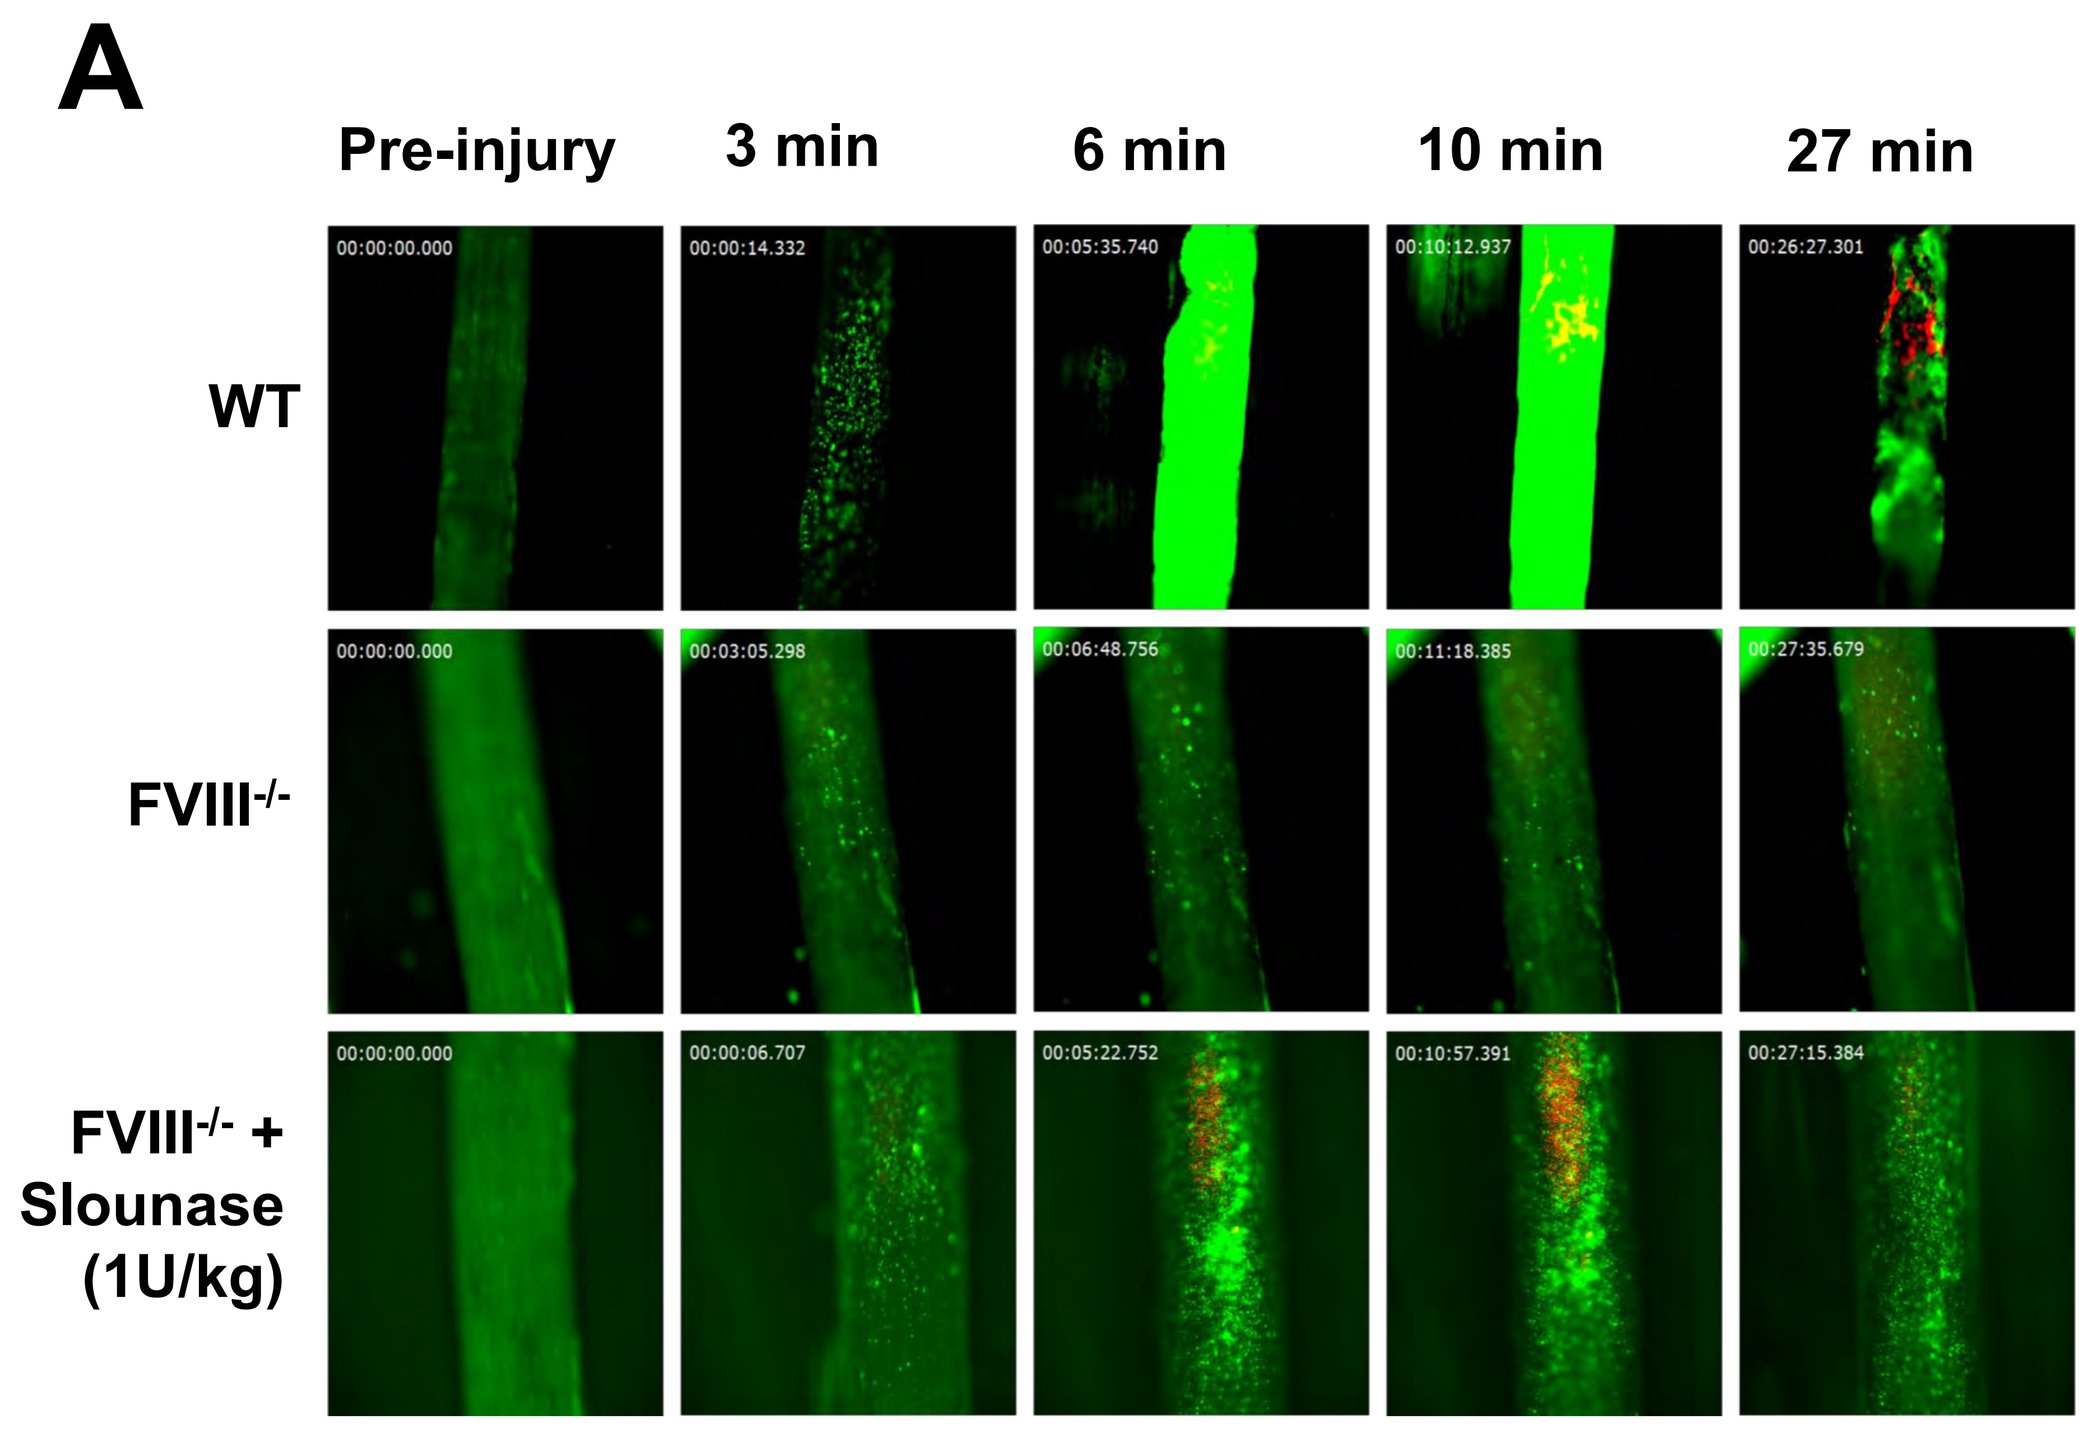
A
Pre-injury
3 min
6 min
10 min
27 min
WT
FVIII-/-
FVIII-/- + Slounase (1U/kg)

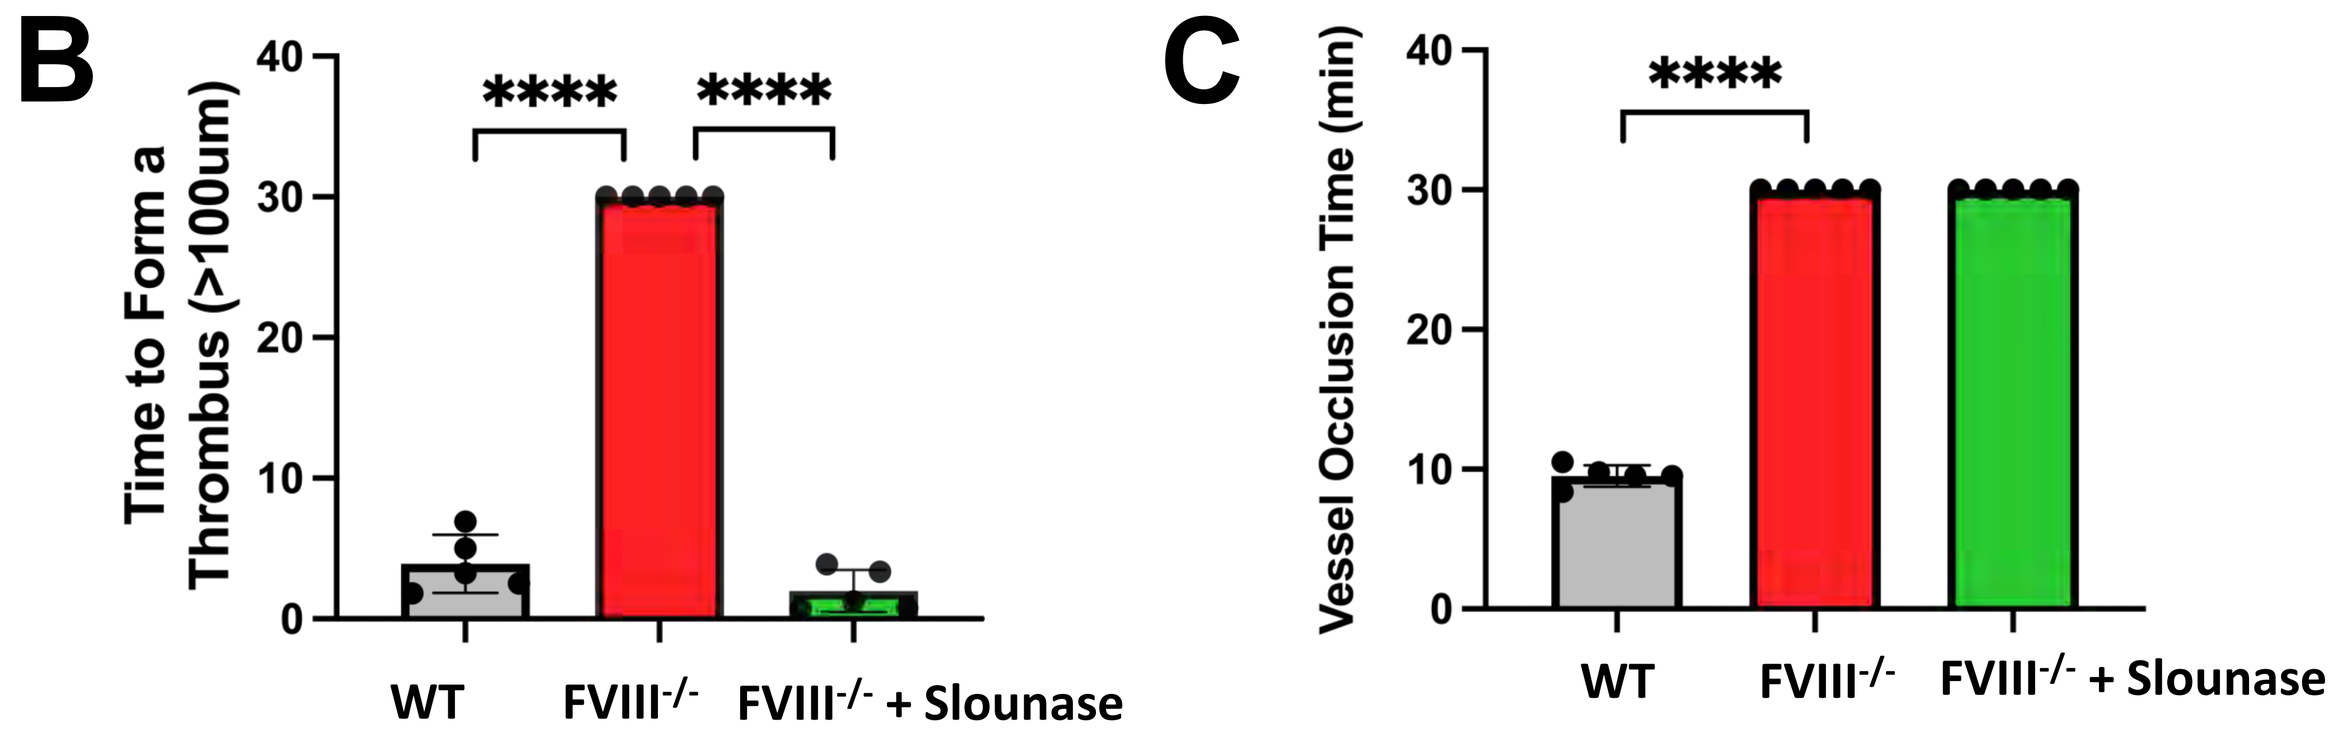
B
Time to Form a Thrombus (>100um)

WT
FVIII-/-
FVIII-/- + Slounase
C
Vessel Occlusion Time (min)

Figure 2: Thrombus formation in response to vascular injury of the carotid artery is severely impaired in FVIII$^{-/-}$ mice and slounase treatment enhanced thrombus formation without reaching vessel occlusion. (A) Representative images of platelet (green) adhesion, aggregation, and fibrin (red) formation within the thrombus after $FeCl_3$-induced carotid artery injury in WT, FVIII$^{-/-}$ mice and FVIII$^{-/-}$ mice pre-treated with slounase. Platelets adhered to the site of vascular injury in the carotid artery in FVIII$^{-/-}$ mice but did not cause formation of large thrombi or vessel occlusion following $FeCl_3$ injury. Prophylactic slounase treatment (1U/kg) in FVIII$^{-/-}$ mice significantly enhanced platelet aggregation and fibrin formation resulting in large stable thrombi without reaching vessel occlusion at the site of vascular injury following $FeCl_3$ injury. **(B)** Time required to form a large thrombus with a diameter more than 100um in WT, FVIII$^{-/-}$ mice and FVIII$^{-/-}$ mice pre-treated with slounase. **(C)** Vessel occlusion time.

Saphenous vein hemostasis model

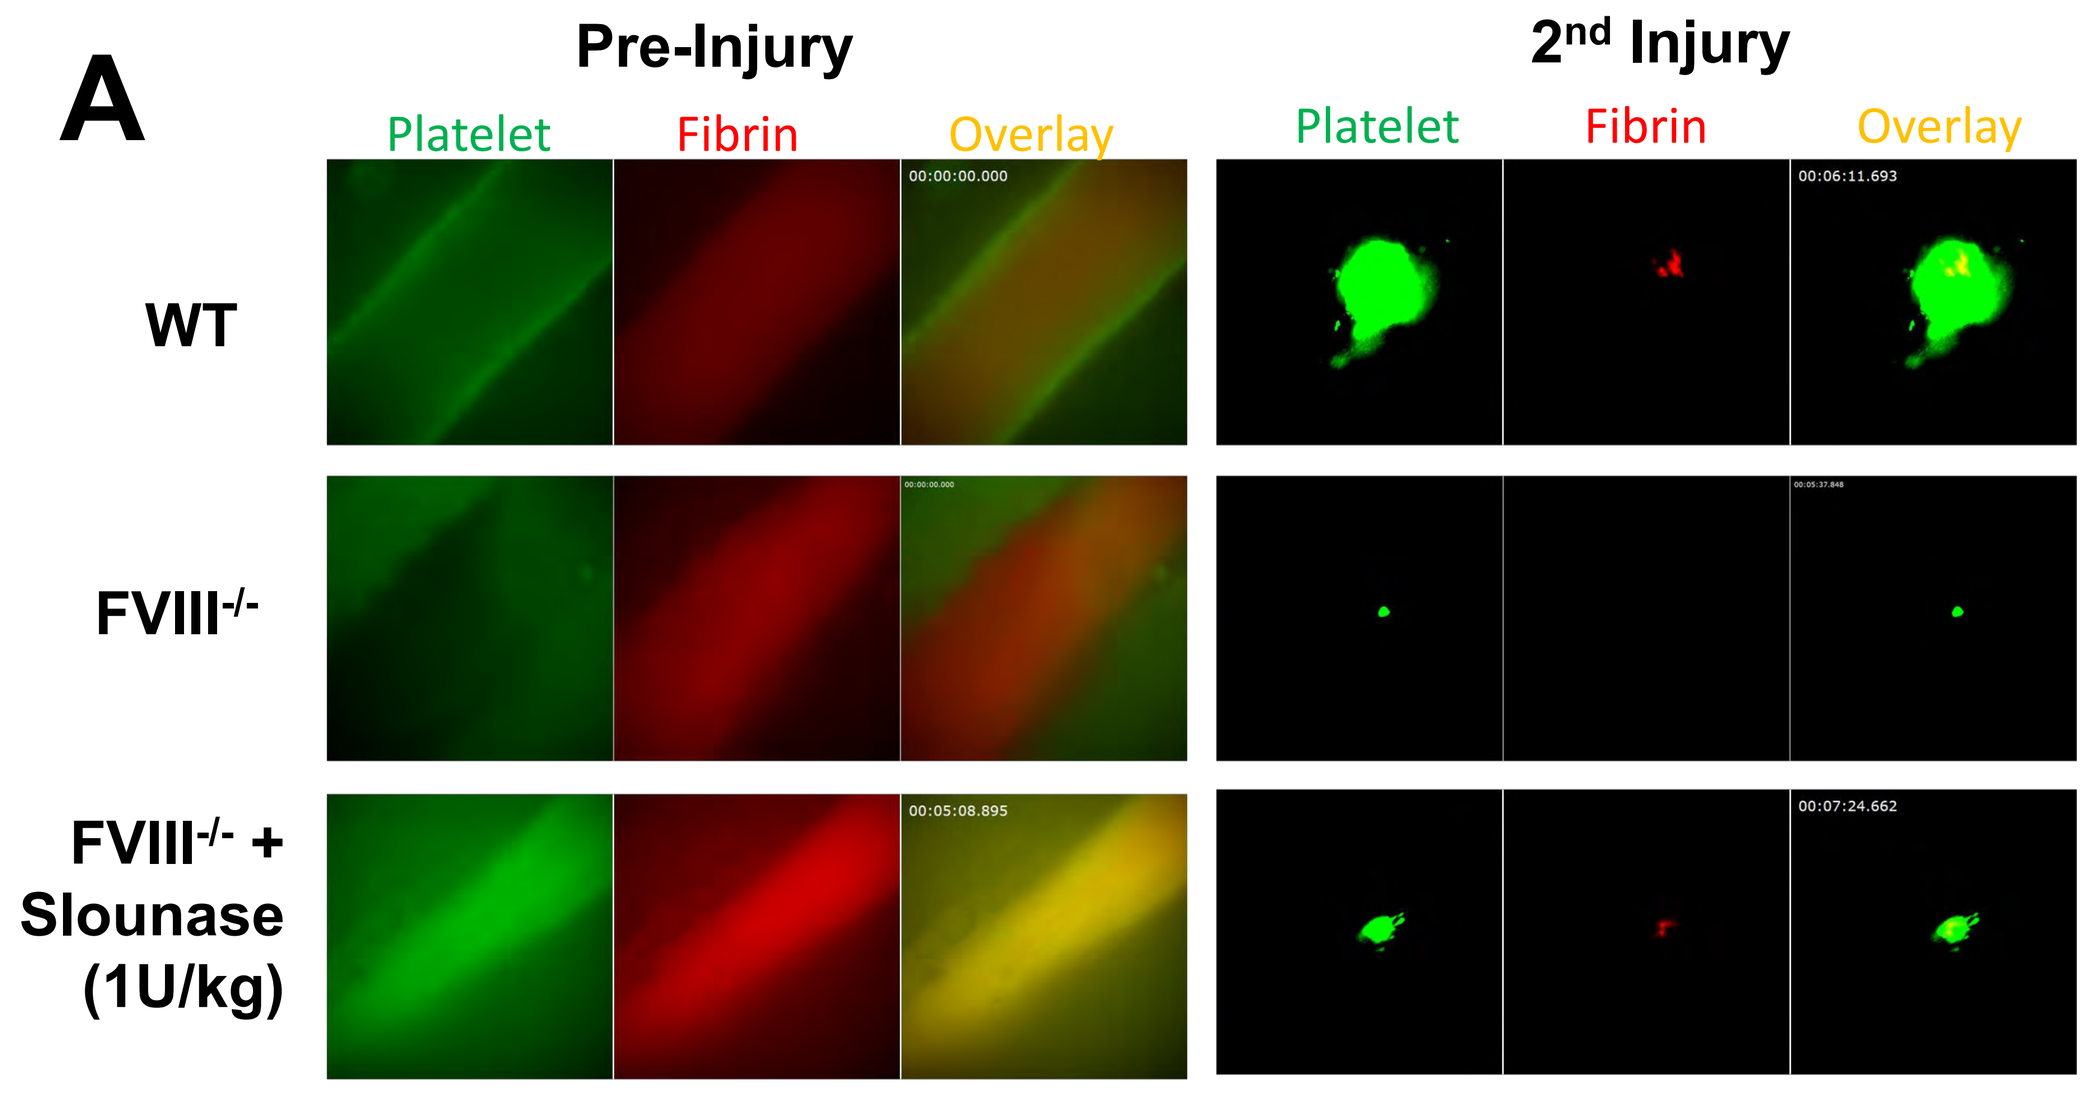
A
Pre-Injury
2nd Injury
Platelet
Fibrin
Overlay
WT
FVIII-/-
FVIII-/- + Slounase (1U/kg)

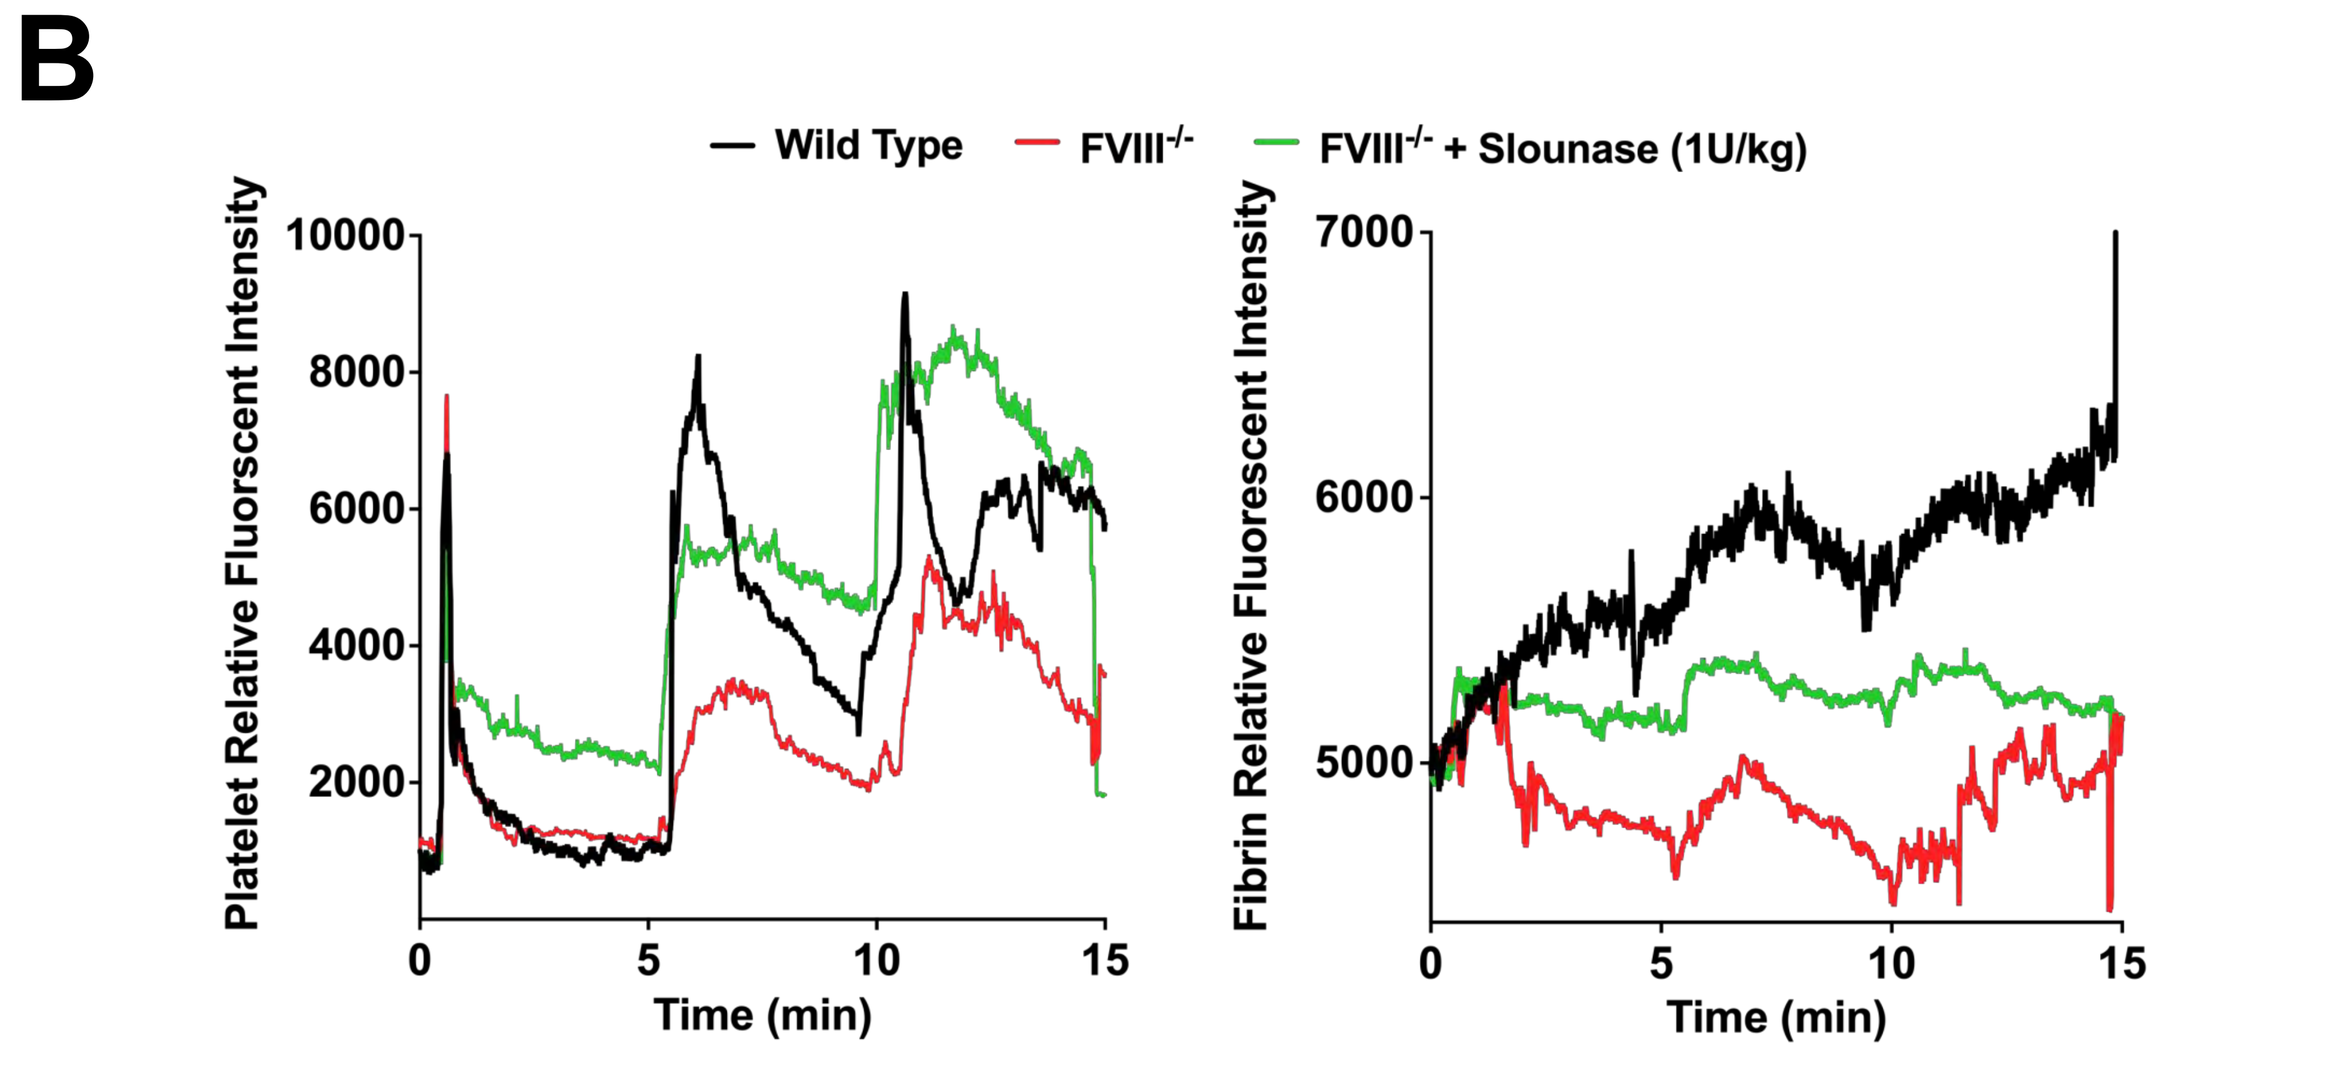
B
Wild Type
FVIII-/-
FVIII-/- + Slounase (1U/kg)
Platelet Relative Fluorescent Intensity
Fibrin Relative Fluorescent Intensity
Time (min)

Figure 3: Slounase treatment improved hemostatic plug formation in FVIII$^{-/-}$ mice as assessed by the saphenous vein rupture model of hemostasis. A penetrative injury on the saphenous vein wall was induced by maximal laser injury at 30 seconds followed by repetitive injuries at 5 min and 10 min and hemostatic plug formation was recorded under intravital microscopy. **(A)** Representative image of platelet accumulation (green) and fibrin formation (red) pre-injury and within the clot after the 2nd injury. **(B)** Quantitative analysis of platelet recruitment (left) and fibrin formation (right) in the thrombus over time. Slounase treatment enhanced thrombus stability, fibrin formation and platelet accumulation in FVIII$^{-/-}$ mice *in vivo*.

SLOUNASE RESTORES HEMOSTATIC PROPERTIES IN FVIII DEFICIENCY *IN VITRO*

Thromboelastography (TEG)

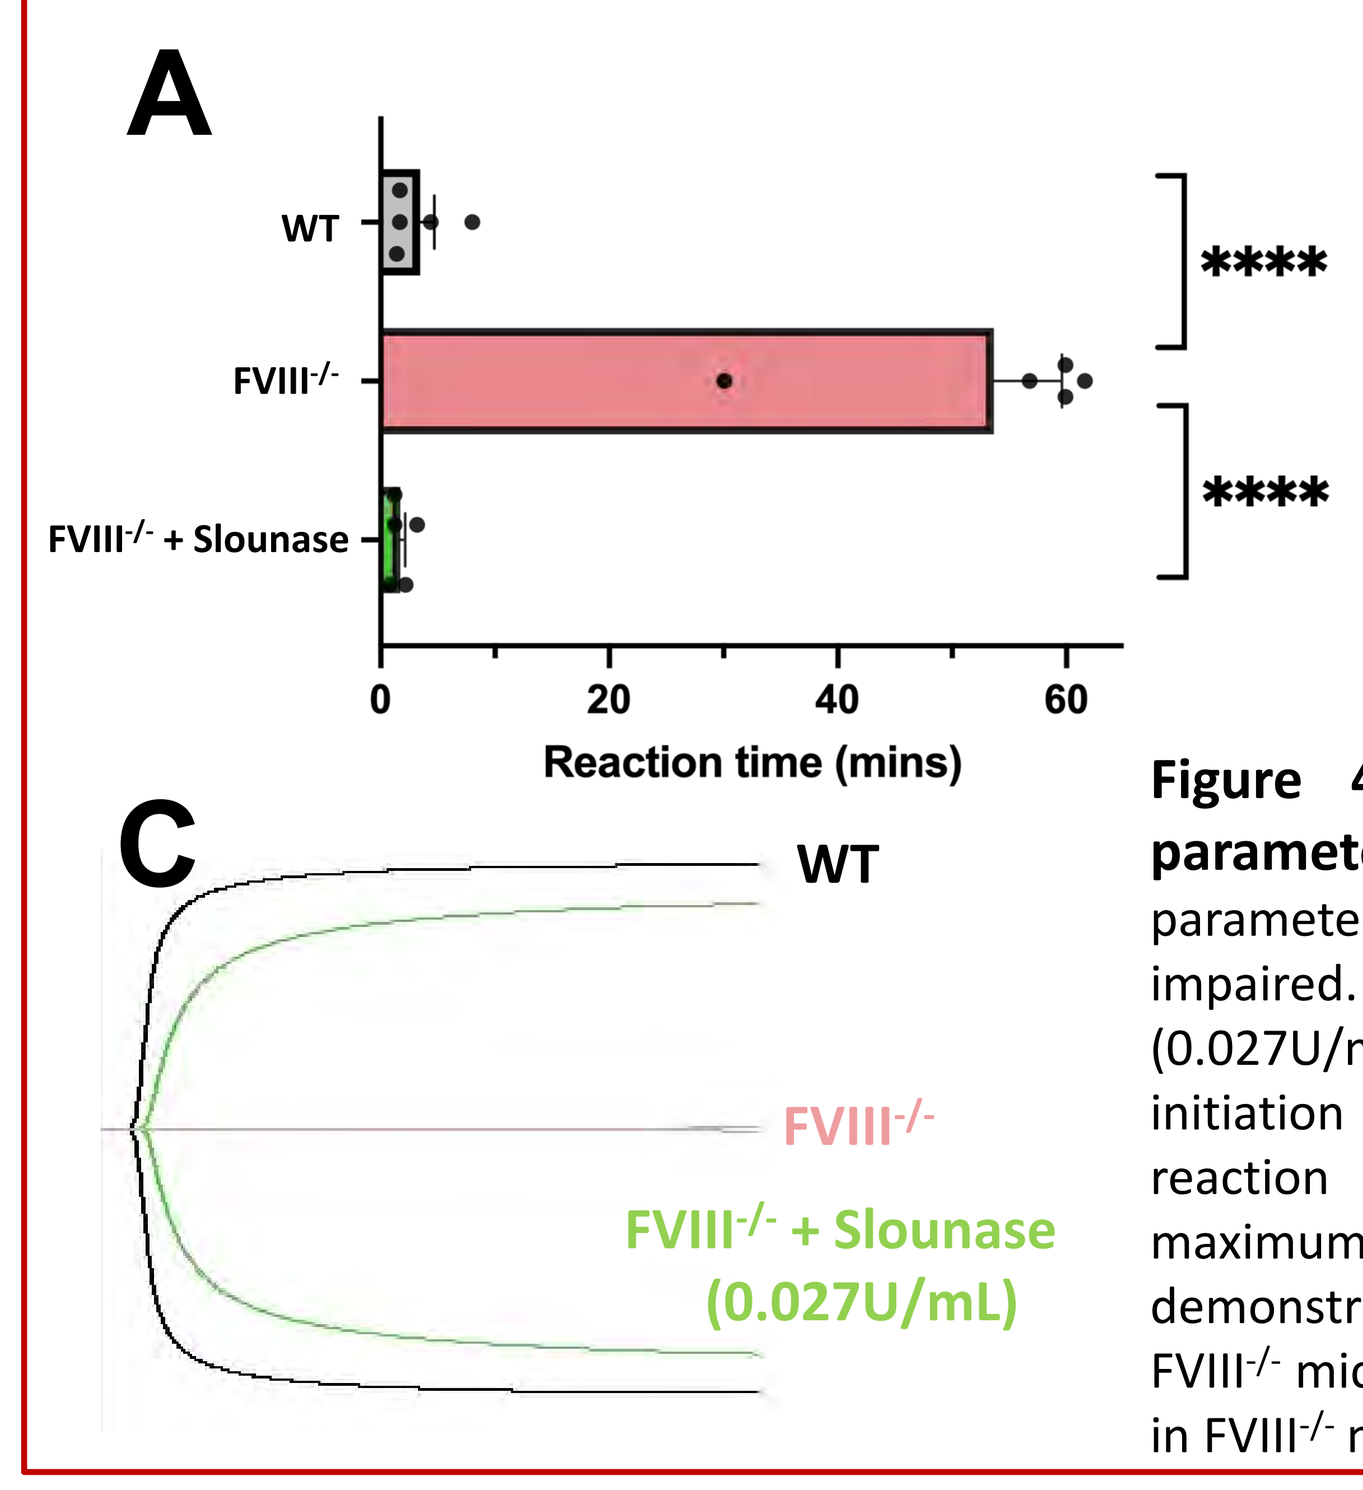
A
WT
FVIII-/-
FVIII-/- + Slounase

Reaction time (mins)
C
WT
FVIII-/-
FVIII-/- + Slounase (0.027U/mL)

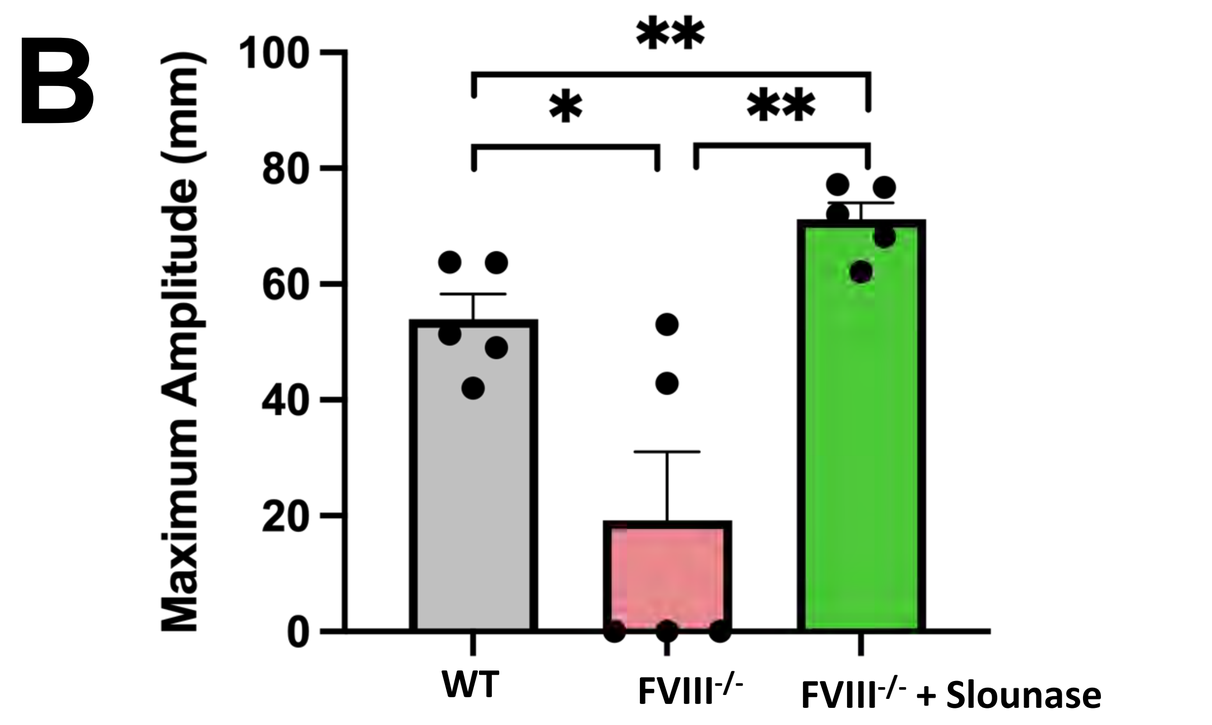
B
Maximum Amplitude (mm)
WT
FVIII-/-
FVIII-/- + Slounase

Figure 4: Slounase treatment restored hemostatic parameters in FVIII$^{-/-}$ mice examined by TEG. Hemostatic parameters in whole blood from FVIII$^{-/-}$ mice were severely impaired. Incubation of FVIII$^{-/-}$ mouse blood with slounase (0.027U/mL) normalized the TEG parameters. **(A)** Rate of clot initiation representing fibrin formation was assessed by reaction time **(B)** Maximal clot strength was assessed by maximum amplitude (MA). **(C)** Representative TEG trace demonstrating the severe impairment of clot formation in FVIII$^{-/-}$ mice and the ability of slounase to restore clot formation in FVIII$^{-/-}$ mice to WT clot formation.

SLOUNASE TREATMENT DECREASES BLOOD LOSS AND BLEEDING TIME IN FVIII$^{-/-}$ MICE

Tail-Bleeding and Hepatic Bleeding

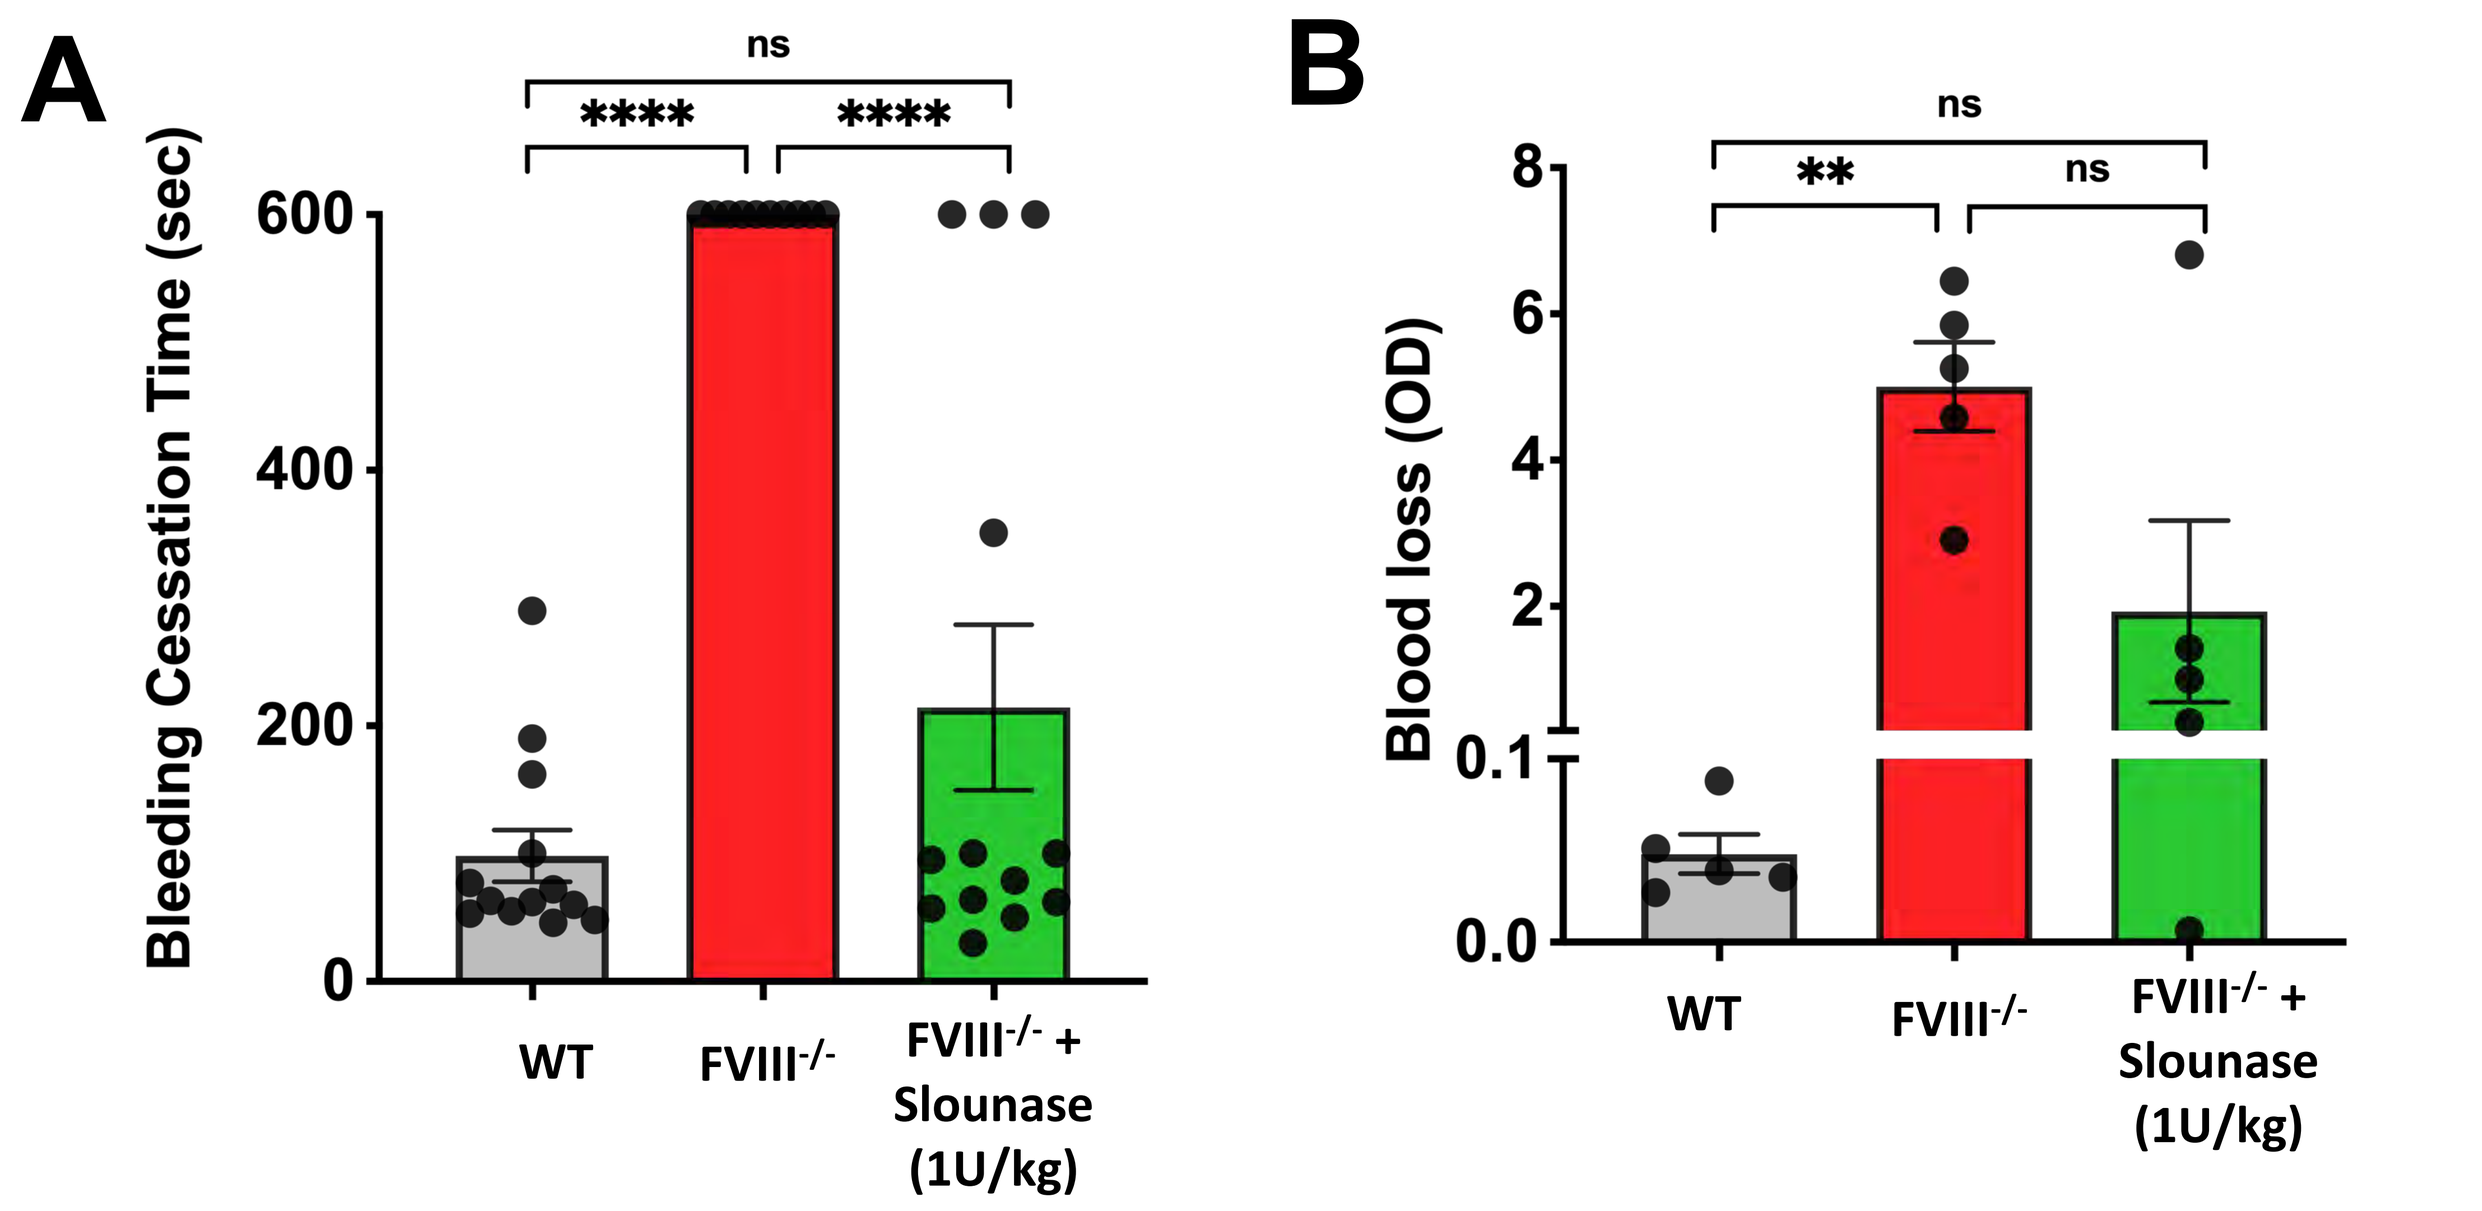
A
Bleeding Cessation Time (sec)
B
Blood loss (OD)
WT
FVIII-/-
FVIII-/- + Slounase (1U/kg)

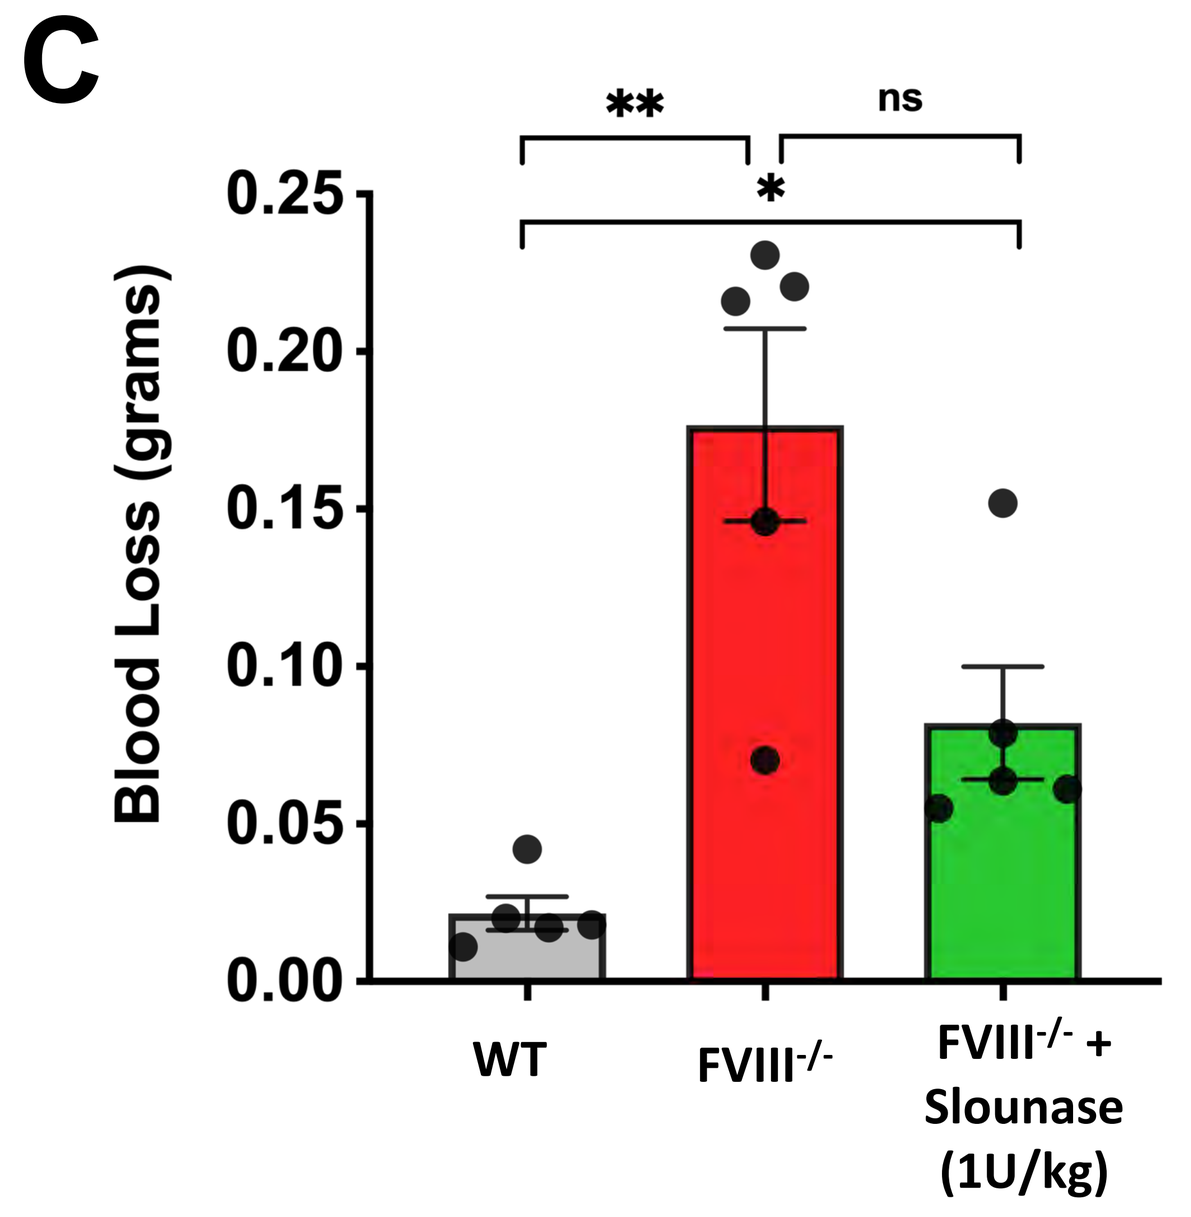
C
Blood Loss (grams)
WT
FVIII-/-
FVIII-/- + Slounase (1U/kg)

Figure 5: Slounase treatment in FVIII$^{-/-}$ mice decreased blood loss and shortened bleeding time. Blood loss and bleeding times were assessed by two different *in vivo* bleeding models in WT, FVIII$^{-/-}$ mice and FVIII$^{-/-}$ mice pretreated with slounase **(A)** Tail bleeding assay: FVIII$^{-/-}$ mice were unable to stop bleeding following tail tip amputation (5mm) and slounase treatment significantly decreased tail bleeding time in FVIII$^{-/-}$ mice. **(B)** Tail tip amputation in FVIII$^{-/-}$ mice resulted in significant blood loss when compared to WT mice as quantified by measuring OD. Slounase treatment decreased the blood loss in FVIII$^{-/-}$ mice. **(C)** Slounase treatment reduced total blood loss in FVIII$^{-/-}$ mice in the *in vivo* liver bleeding model. A penetrative injury on the left liver lobe was induced using a 16G needle and total blood loss was assessed.

CONCLUSIONS

Our data indicates slounase is a novel bypassing agent that promotes platelet procoagulant activity and fibrin formation, to restore hemostasis and limit bleeding in hemophilia A, with and without inhibitors.